PRECLINICAL *IN VITRO* AND *IN VIVO* EVALUATION OF CD8αβ+ CD19 CAR IPSC T CELLS GENERATED IN A TCR SIGNAL INDEPENDENT MANNER USING DLL4/VCAM-COUPLED MICROBEADS

Muluken S. Belew[1], Richard Carpenedo[1], Alessia Pallaoro[1], Hiofan Hoi[1], Avisek Deyati[2], Daniel C. Kirouac[2], Siddarth Chandrasekaran[2], Valerie Wall[2], Libin Abraham[2], Sommer Apelu[2], Michael Hunter Cadell[2], Amanda AuYeung[1], Omar Subedar[1], Elisa Martinez[1], Shri Joshi[1], Elizabeth Csaszar[1], Steven Woodside[1], Emily Titus[1], Christopher Bond[2]

Notch THERAPEUTICS

AACR Annual Meeting 2024 | Poster #39
1 Notch Therapeutics Inc, Toronto, ON, Canada
2 Notch Therapeutics Inc, Seattle, WA, USA

notchtx.com

OVERVIEW

The use of clonally-derived, induced pluripotent stem cells (iPSCs) as starting material for therapeutic T cell manufacturing would overcome many limitations of autologous Chimeric Antigen Receptor T cell (CAR-T) therapies. Complex, multi-stage genome engineering, large batch production and rigorous lot testing could provide a consistent source of off-the-shelf, functionally enhanced T cell products. However, this vision cannot be realized using existing T cell differentiation platforms, which fail to present Notch ligands with the precision and intensity required to control T cell differentiation in scalable, suspension culture.

We have previously shown that VCAM1 synergizes with DLL4 to enhance Notch signaling and progenitor T cell differentiation. We have extended the utility of this discovery through the creation of the Engineered Thymic Niche (ETN) system composed of DLL4/VCAM1-coated magnetic microbeads that provides precise and tunable control of Notch signaling intensity and duration in a scalable, suspension culture format.

Using a clonal iPSC line with a CD19 CAR knocked in at the TRAC locus, we apply this approach to generate CAR-expressing, CD8αβ+ functional T cells which are transcriptionally similar to primary CD8+ T cells and capable of multiple rounds of *in vitro* tumor cell lysis as well as sustained tumor growth inhibition *in vivo*. This advancement demonstrates proof-of-concept for generation of highly efficacious, off-the-shelf CAR-T cells using novel, small-footprint manufacturing in order to broaden the applicability and accessibility of T cell therapies for cancer patients.

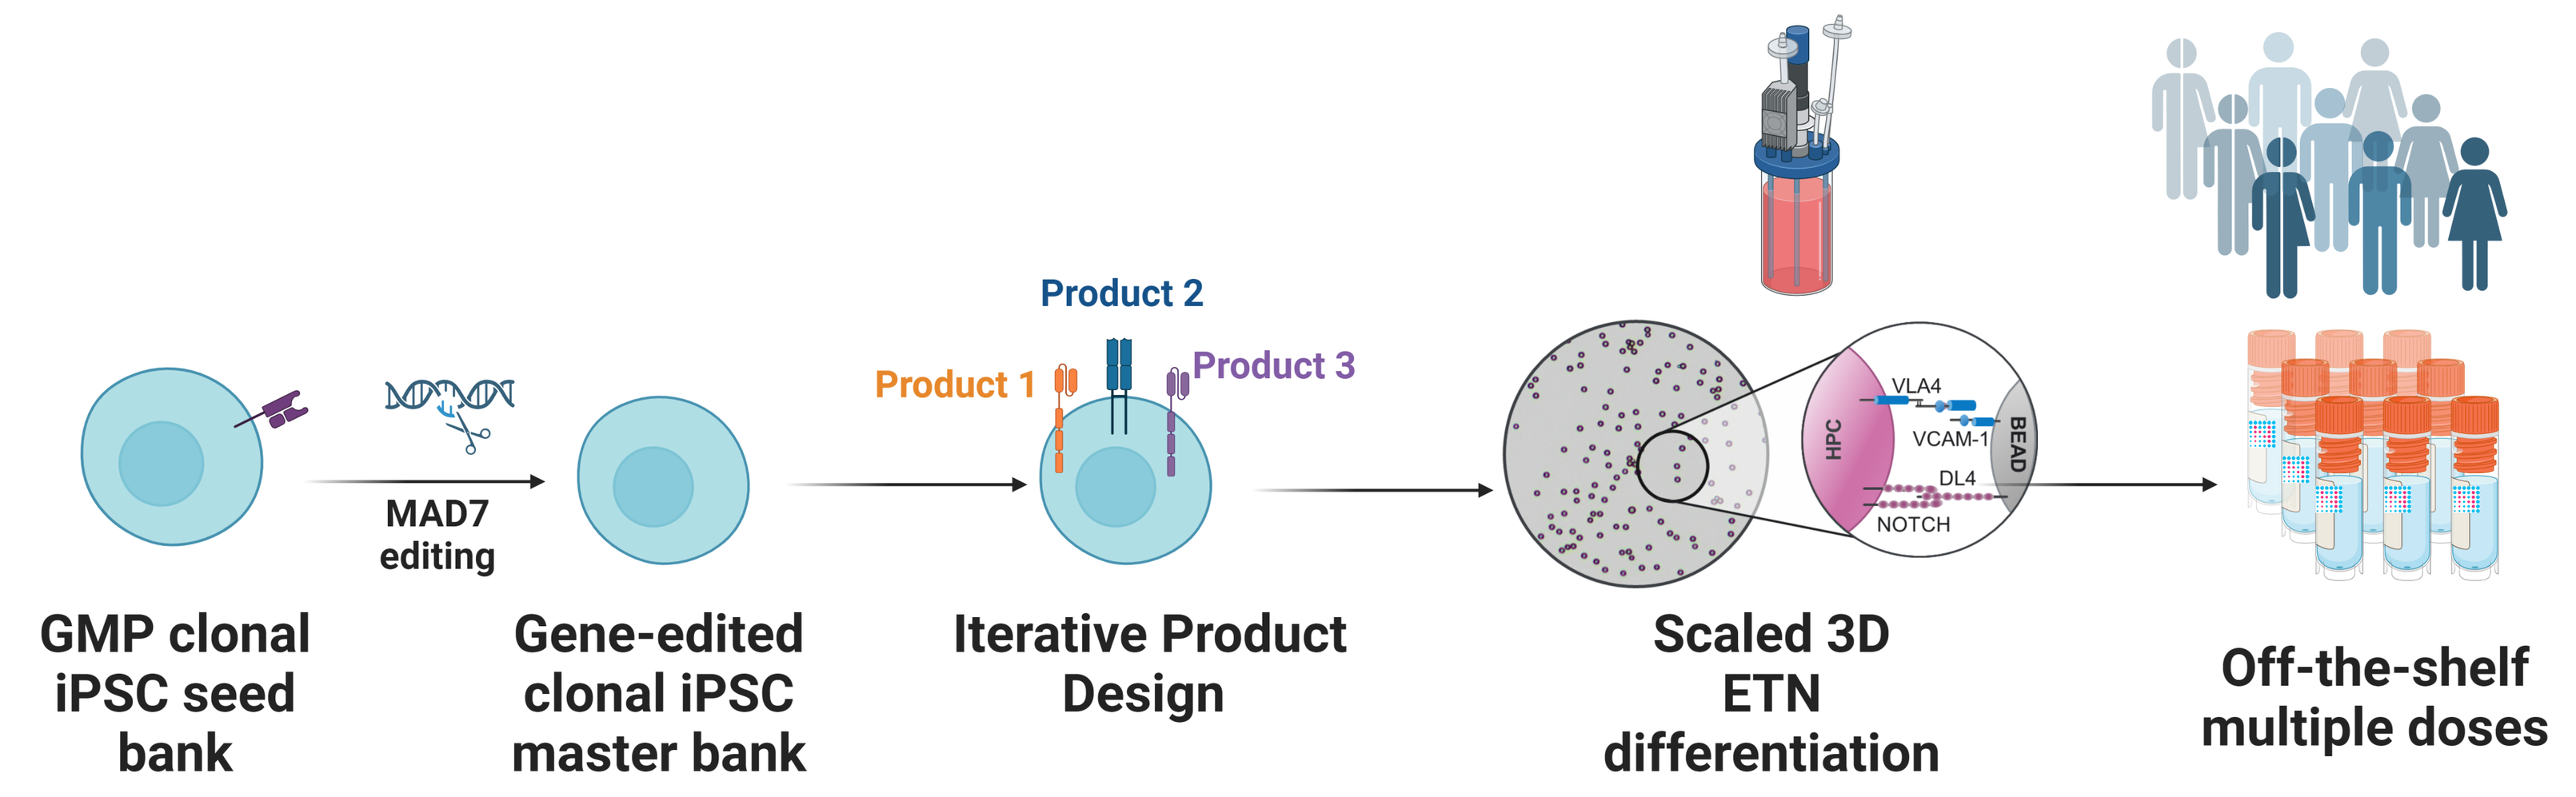

MAD7 editing
Product 1
Product 2
Product 3
VLA4
VCAM-1
DL4
NOTCH
HPC
BEAD
GMP clonal iPSC seed bank
Gene-edited clonal iPSC master bank
Iterative Product Design
Scaled 3D ETN differentiation
Off-the-shelf multiple doses

SUMMARY OF METHODS

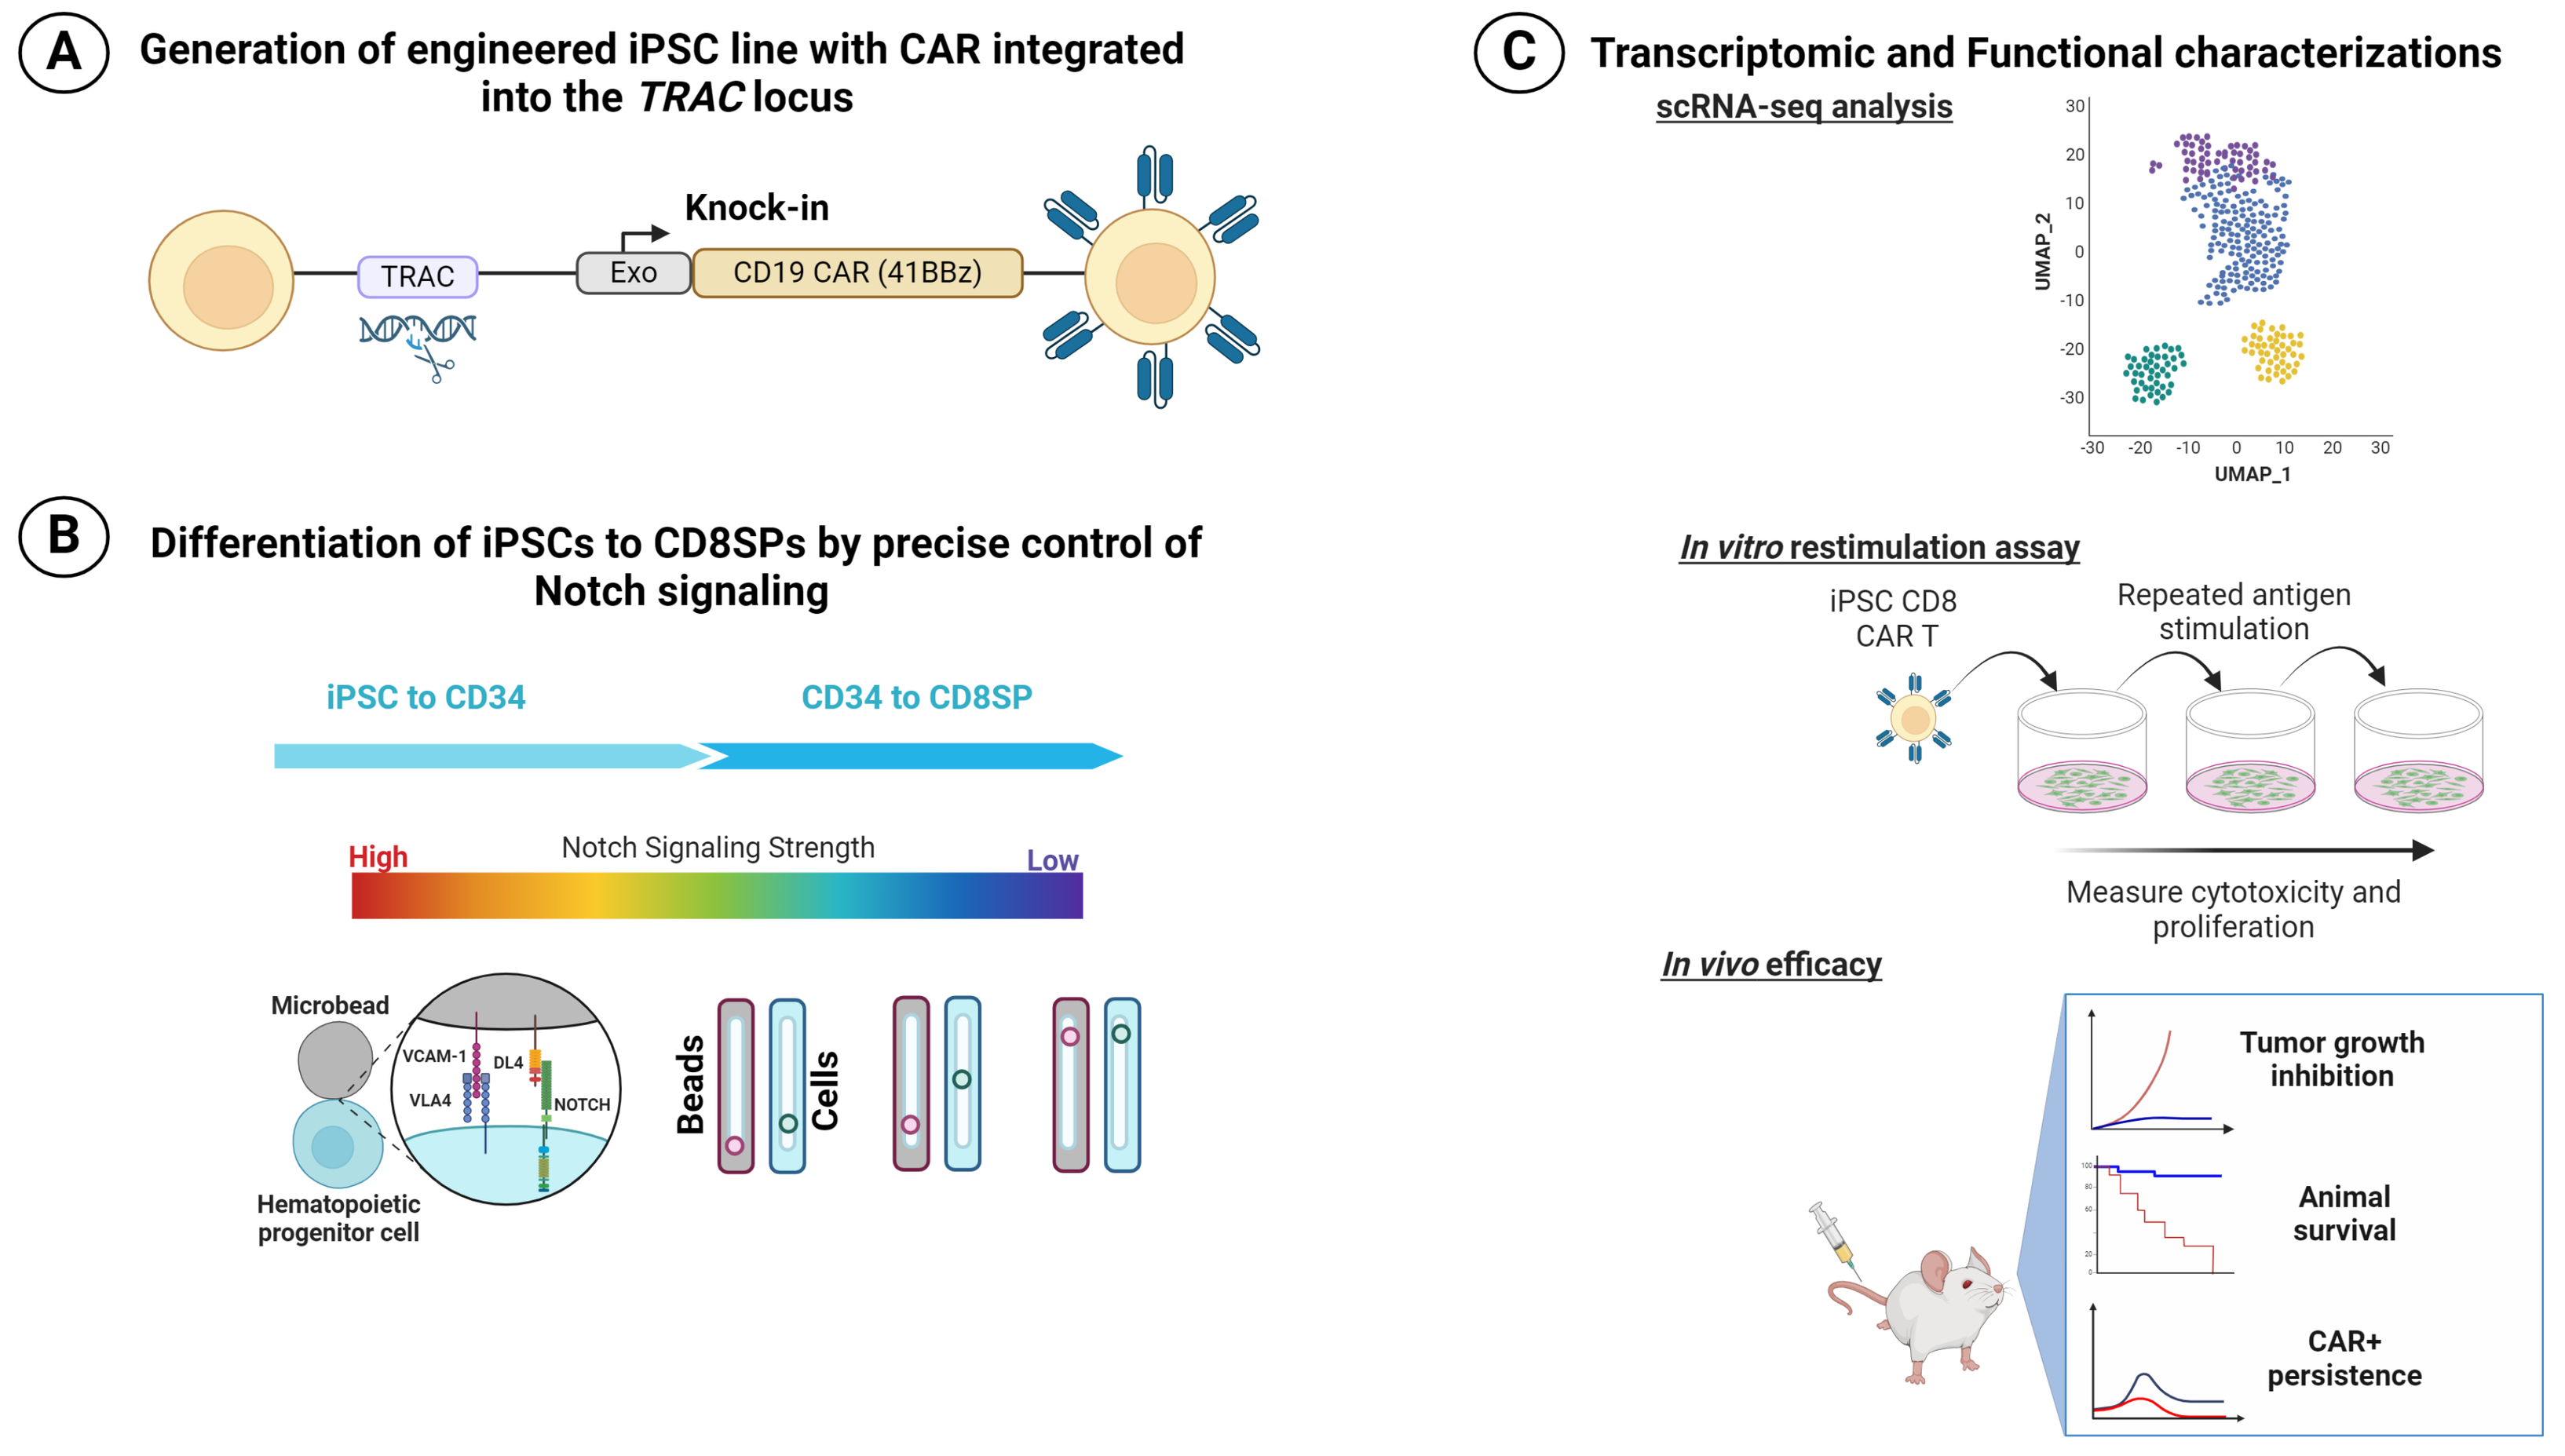

A Generation of engineered iPSC line with CAR integrated into the TRAC locus
TRAC
Knock-in
Exo
CD19 CAR (41BBz)
B Differentiation of iPSCs to CD8SPs by precise control of Notch signaling
iPSC to CD34
CD34 to CD8SP
High
Notch Signaling Strength
Low
Microbead
VCAM-1
DL4
VLA4
NOTCH
Hematopoietic progenitor cell
Beads
Cells
C Transcriptomic and Functional characterizations
scRNA-seq analysis
UMAP_1
UMAP_2
In vitro restimulation assay
iPSC CD8 CAR T
Repeated antigen stimulation
Measure cytotoxicity and proliferation
In vivo efficacy
Tumor growth inhibition
Animal survival
CAR+ persistence

A. IPSCs were engineered to be TRAC-/- with site-specific integration of a CD19 CAR (41BBz) under exogenous promotor control into the TRAC Locus.

B. Engineered iPSCs were differentiated to CD34+ hematopoietic progenitor cells before undergoing differentiation to CD8+ T cells in a suspension culture via precise control of Notch signaling in the presence of ETN beads.

C. IPSC-derived CD8+ T cell phenotype was characterized using flow cytometry and transcriptomic analysis. Antigen-mediated cytotoxicity and T cell proliferation were assessed *in vitro* using live cell imaging of iPSC-T cells co-cultured via serial restimulation with CD19-expressing tumor cells. An *in vivo* efficacy study was conducted in NSG mice engrafted with a disseminated A549 CD19-expressing tumor cell line, with bi-weekly readouts of tumor growth inhibition, animal survival, and body weight, and weekly readouts of CAR-T persistence.

RESULTS

Figure 1. CD8αβ+ T cells are generated from iPSC-derived hematopoietic progenitor cells using DLL4/VCAM-1 microbeads

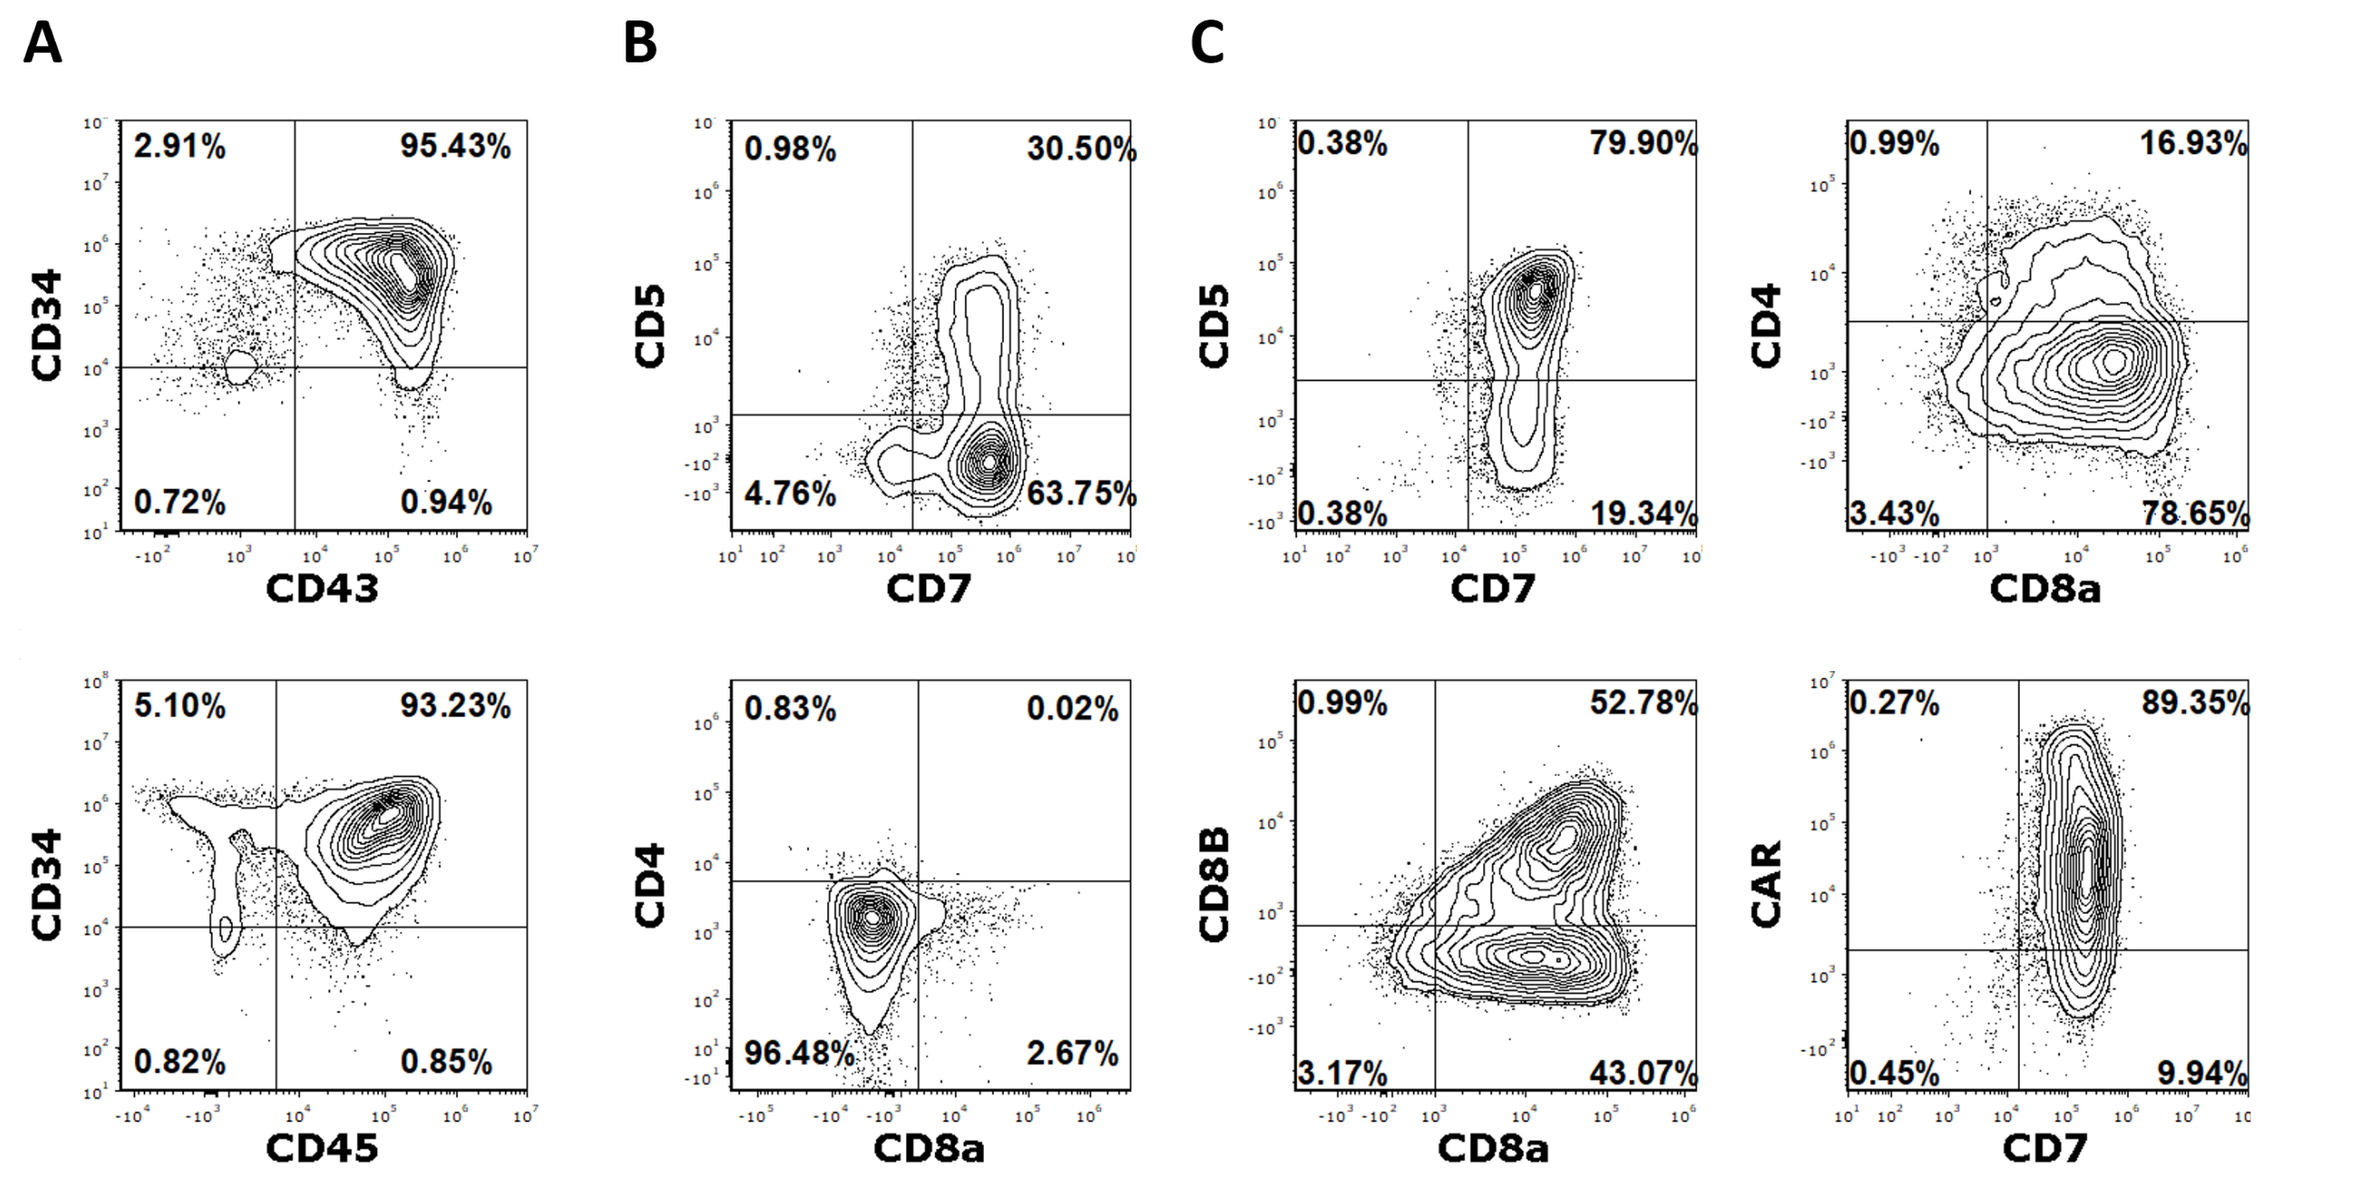

A
B
C
CD34
CD43
CD45
CD5
CD7
CD4
CD8a
CD8B
CAR

D

End of process Parameters	%
Viability	92
CD5+CD7+	80
CD7+	100
CD8α+	95
CDαβ+	52
CAR+	90

A. At the initiation of lymphoid induction (day 0), cells were primarily CD34+/CD43+/CD45+ (>90%).

B. Following 10 days of differentiation, cells were lymphoid committed (95% CD7+, and ~30% CD5/7+) and did not yet express CD4 and CD8 T cell markers.

C. By day 21, cells had committed to a CD8αβ cytotoxic T cell phenotype, subjected to CD8 enrichment, resulting in a population of CD8+ T cells with high CD19 CAR expression.

D. Summary of key phenotypes at the end of differentiation process. While cumulative fold expansion and viability vary throughout the differentiation process, markers for T-lineage commitment increase throughout the 21-day time course.

Figure 2: Activated iPSC CD19 CAR T cells transcriptionally cluster closely with activated PBMC-derived CAR T cells

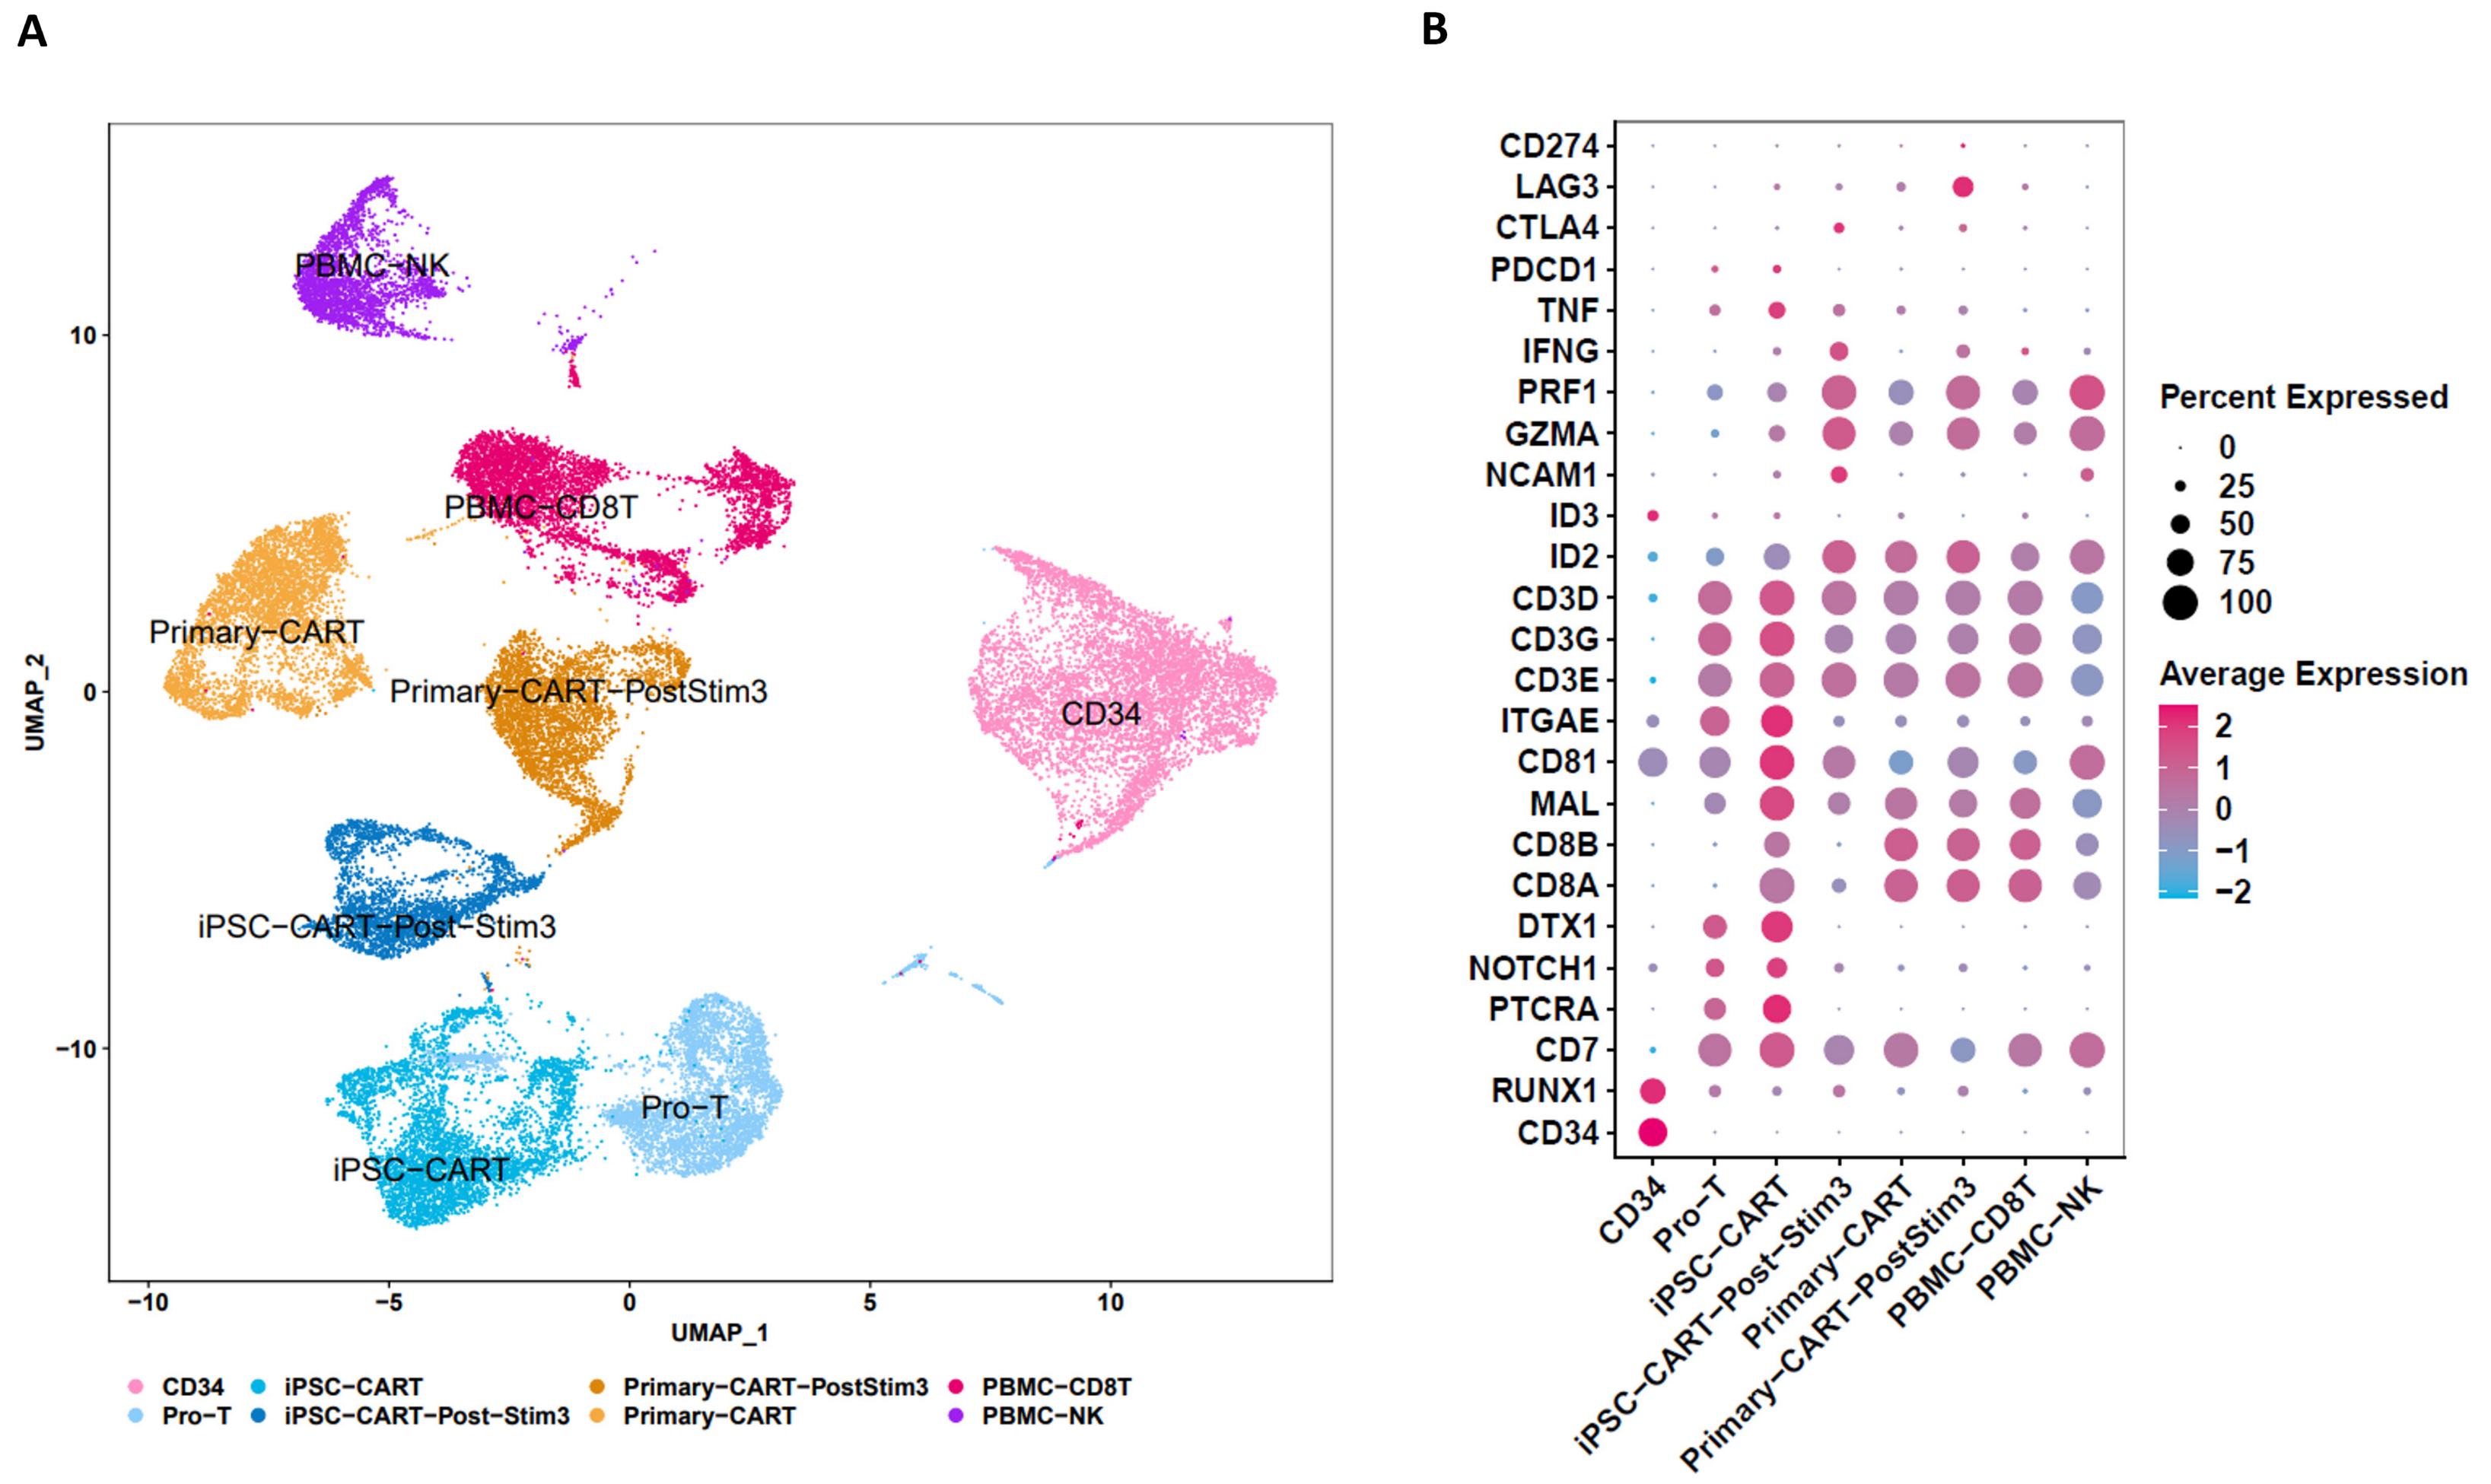

A
B
UMAP_1
UMAP_2
PBMC-NK
PBMC-CD8T
Primary-CART
Primary-CART-PostStim3
CD34
iPSC-CART-Post-Stim3
iPSC-CART
Pro-T
Percent Expressed
Average Expression

A. UMAP of single cell transcriptomes; iPSC-derived CAR T cells through differentiation (CD34+, Pro-T, CD8+SP stage, and following three rounds of exposure to A549 CD19+/+ tumor cell stimulation) vs. PBMCs (CD8+ T cells, CD56+ NK cells). Analysis also included primary CD19 CAR T cells following three rounds of antigen stimulations. The transcriptomes of iPSC CAR T cells and PBMC-derived CAR T cells align closely after restimulation assays with antigen-expressing tumor cells.

B. A bubble plot of select genes associated with blood progenitors, T cell lineage commitment, canonical T cell function, innate lymphocytes (ILCs), cytotoxic T cells, exhausted T cells, and Notch response elements. iPSC-derived CD8+ T cells acquire a canonical T cell gene expression pattern during differentiation, with heightened CD3 expression, limited expression of ILC or exhausted markers, and activated Notch signaling. Upon CD19 antigen stimulation, iPSC-derived CAR T cells demonstrate heightened Granzyme and Perforin expression as expected.

Figure 3: IPSC-derived CD19 CAR-T cells demonstrate robust *in vitro* cytotoxicity and proliferation

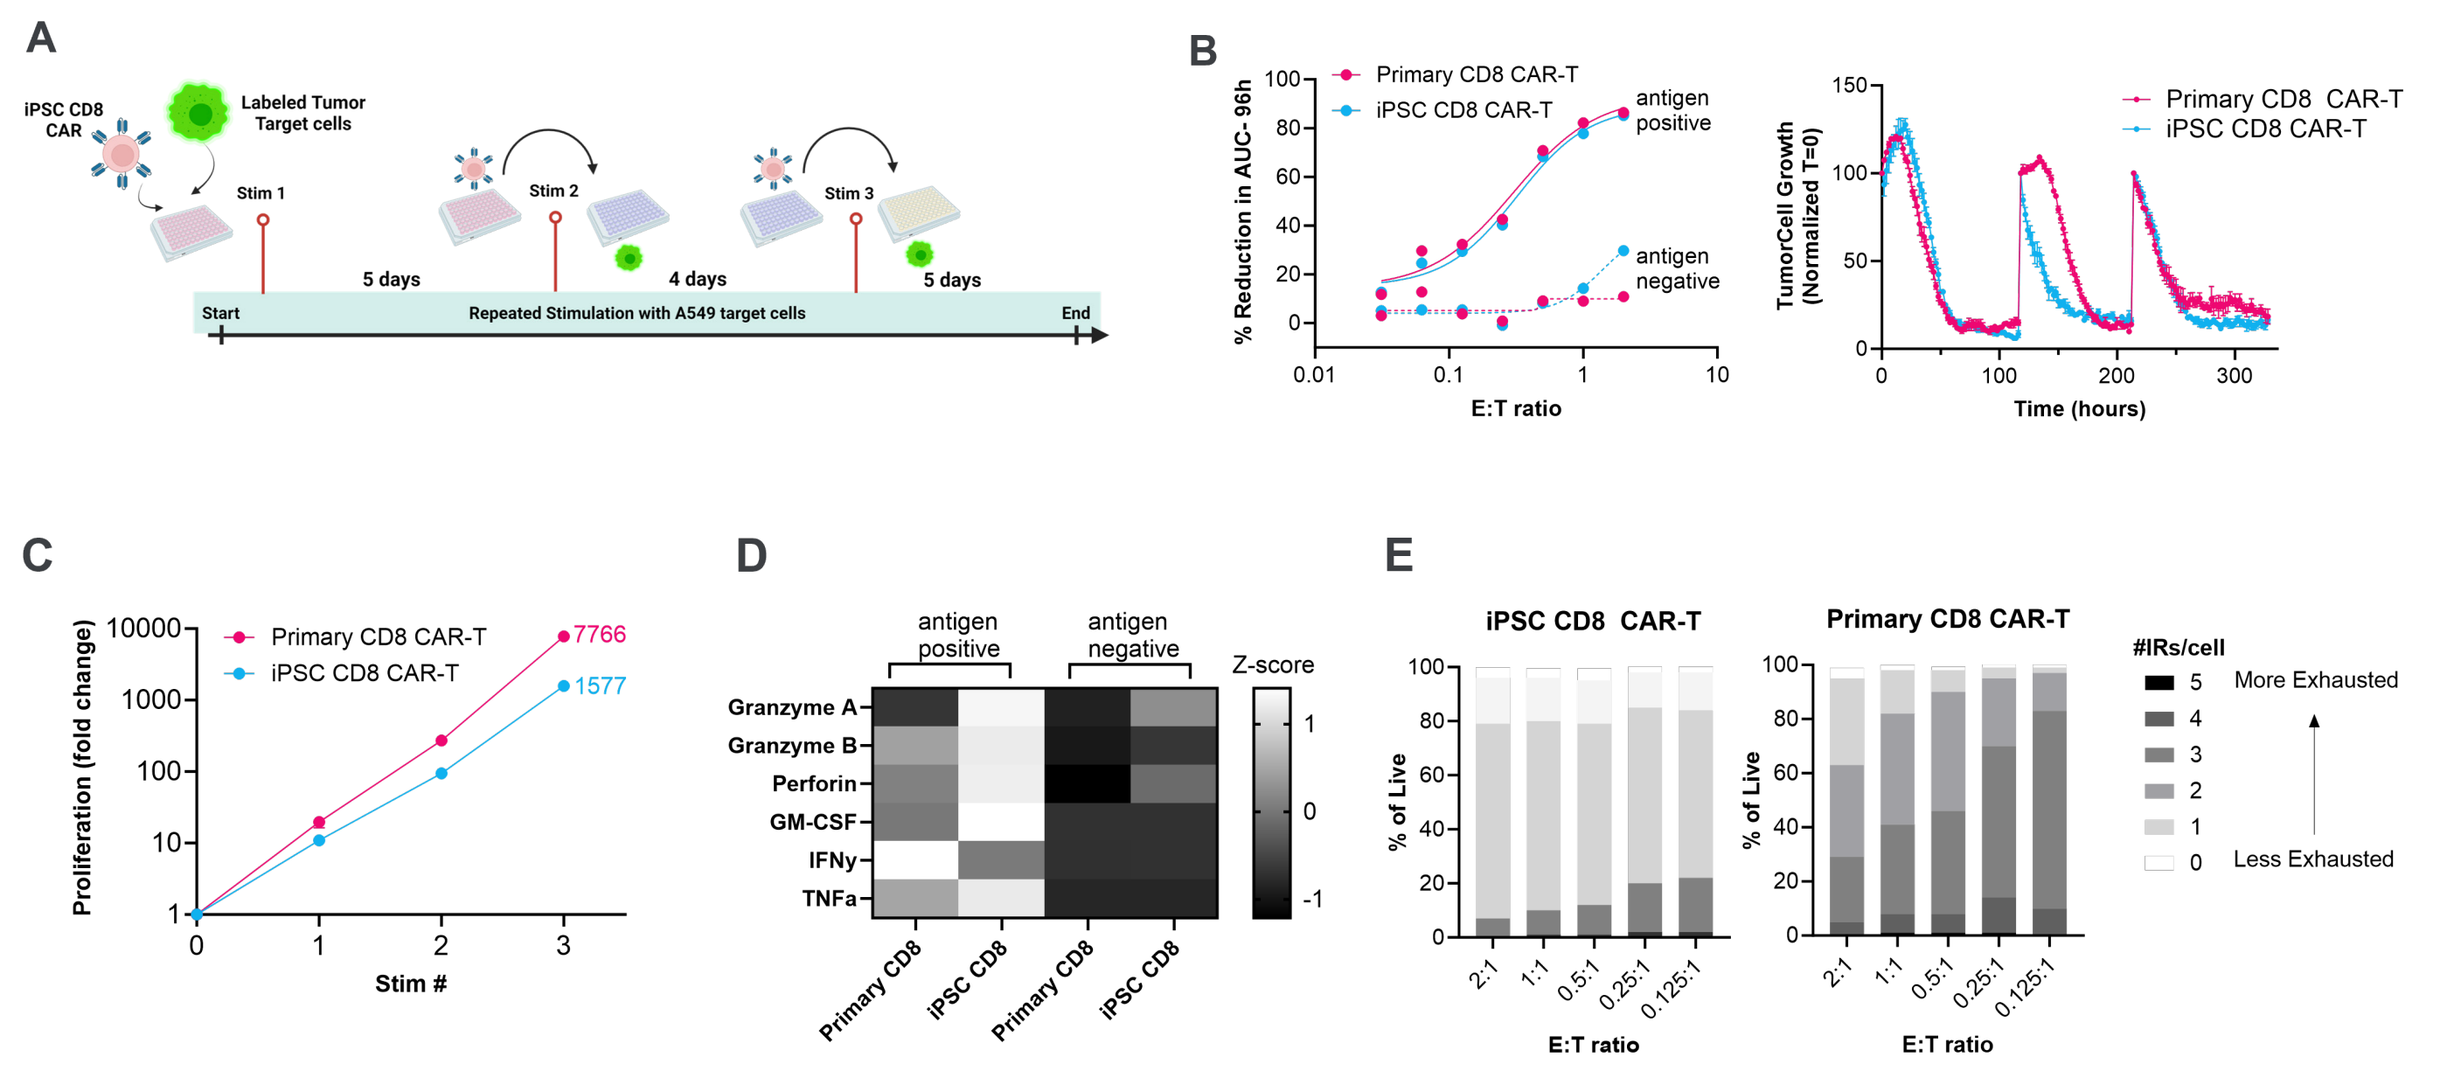

A
B
C
D
E
Primary CD8 CAR-T
iPSC CD8 CAR-T
E:T ratio
Time (hours)
Stim #
antigen positive
antigen negative
Z-score
iPSC CD8 CAR-T
Primary CD8 CAR-T
More Exhausted
Less Exhausted

A. Serial restimulation assay schematic. iPSC CD8 CAR-T or Primary CD8 CAR-T (effector cells) are seeded with NuclightTM green-labeled A549 CD19+/+ tumor targets at multiple E:T ratios and co-cultured for 5 days in an Incucyte to monitor cytotoxicity (Stim 1). Following harvest at day 5, effector cells from the 2:1 E:T ratio are counted and reseeded at a 2:1 E:T for a subsequent stimulation with new tumor targets. This process was repeated for a total of 3 rounds of activity (14 days total).

B. Using an Incucyte to monitor target cell killing, cells were challenged for a single round of activation at progressively lower E:T ratios. iPSC CD8 T cells demonstrated target-specific cytotoxicity comparable to that of Primary CD8 CAR-Ts for all E:T ratios tested (left). Using an optimal E:T ratio for iPSC CD8 (2:1), iPSC CD8 CAR-Ts demonstrate repeated killing of tumor targets comparable to Primary CD8 CAR-Ts (right).

C. Proliferation was assessed by quantification of number of T cells per well before and after each round of activation allowing for calculation of fold change. iPSC CD8 CAR-Ts continue to proliferate during each round of stimulation.

D. Cytokine secretion following 48h of culture with antigen positive or antigen negative tumor cells was assessed by MSD. iPSC CD8 CAR-Ts exhibit target-specific secretion of cytokines.

E. T cell phenotype was monitored by flow cytometry throughout the restimulation assay at the multiple E:T ratios. Lower E:T ratios result in higher co-expression of inhibitory receptors (IRs; PD-1, TIGIT, LAG3, TIM3 and CD39) on effector cells (a measure of CD8 T cell exhaustion).

Figure 4: IPSC-derived CD19 CAR T cells demonstrate persistent *in vivo* efficacy in xenograft NSG mouse tumor model

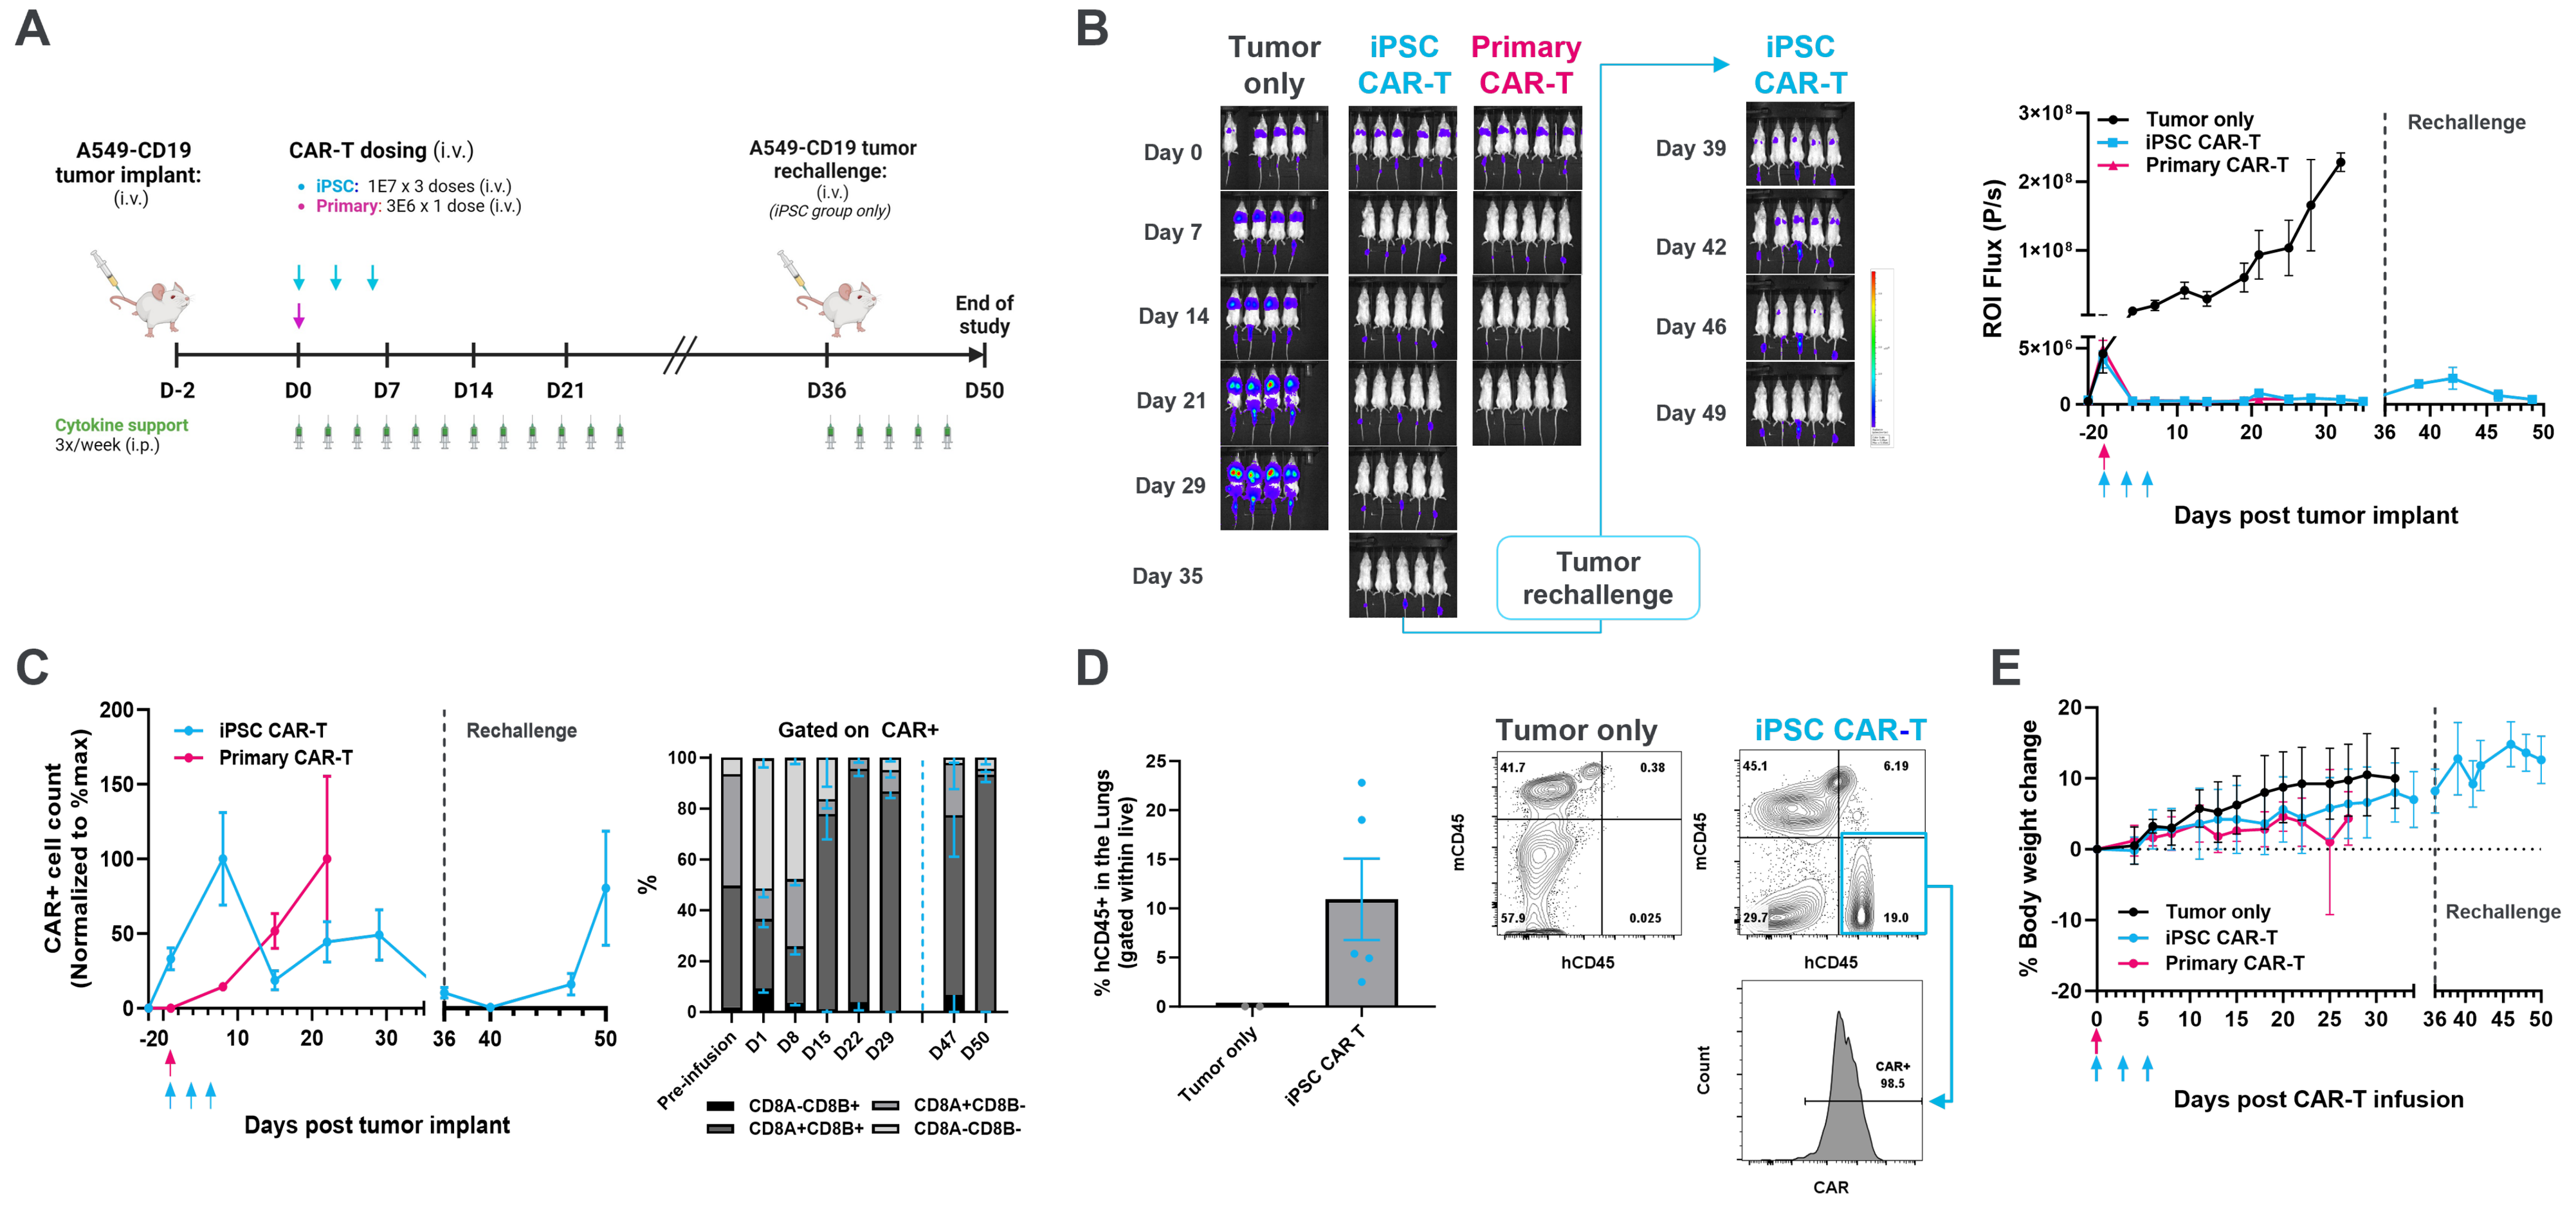

A
B
C
D
E
A549-CD19 tumor implant: (i.v.)
CAR-T dosing (i.v.)
iPSC: 1E7 x 3 doses (i.v.)
Primary: 3E6 x 1 dose (i.v.)
A549-CD19 tumor rechallenge: (i.v.) (iPSC group only)
End of study
Cytokine support 3x/week (i.p.)
Tumor only
iPSC CAR-T
Primary CAR-T
Tumor rechallenge
Rechallenge
Days post tumor implant
Gated on CAR+
Days post CAR-T infusion

A. Schematic of treatment regimen for NSG mice bearing disseminated A549-CD19 tumor model. A549 CD19+/+ cells co-expressing luciferase were implanted into 6-7 week old NSG mice through lateral tail vein injection, two days before first CAR T injections. A total of 3x107 iPSC CAR+ CD8+ cells were administered intravenously in 3 doses. A single 3x106 dose of CD8+ enriched primary CAR-Ts was used as a positive control. Study cohorts received cytokine support intraperitoneally 3 times per week for 4 weeks. On Day 36, mice treated with the iPSC CAR-T cells were rechallenged with additional A549 CD19+/+ cells injected through the lateral tail vein. Cytokine support was provided intraperitoneally 3 times per week for an additional 2 weeks.

B. Tumor burden was monitored bi-weekly by bioluminescence imaging using IVIS Lumina III through intraperitoneal injection of D-luciferin substrate in sterile Phosphate-buffered saline. Images of representative time points are shown before (left panel) and after (middle panel) tumor rechallenge. Quantification of tumor burden as region of interest (ROI) flux values are shown in the right panel with each dot representing mean of the treatment cohorts and error bars represent SEM.

C. CAR+ cell persistence was evaluated by flow cytometry from peripheral blood (left panel) and phenotypic analysis of circulating CAR+ cells (right panel) before and after tumor rechallenge.

D. End of life analysis by flow cytometry of iPSC-derived CAR T cells after mechanical dissociation of the lungs to single cell suspensions was performed. Right panel indicates representative flow plots.

E. Percent change in body weight was monitored throughout study for assessment of acute toxicity.

CONCLUSIONS

- Controlled Notch signaling provided via the Engineered Thymic Niche technology enables generation of iPSC-derived CD8+ T cells, bypassing the necessity for TCR/CD3 signaling and enabling allogeneic T cell generation for off-the-shelf therapy development.
- Using a clonal, TCR-deficient iPSC cell line with a CD19 CAR knocked in at the TRAC locus, we have applied this approach to generate iPSC-CAR T cells, capable of multiple rounds of *in vitro* tumor cell lysis and sustained tumor growth inhibition *in vivo* comparable to primary T cells and without acute toxicity.
- This advancement demonstrates proof-of-concept for generation of highly efficacious, off-the-shelf CAR-T cells using novel, small-footprint manufacturing in order to broaden the applicability and accessibility of T cell therapies for cancer patients.